Use of High Titer RSV Immunoglobulin (RI-001-RSV IVIG) in Immunocompromised Adults

A FALSEY, C KOVAL, M KHORANA, E WALSH, M SORRENTINO, R MURRAY, R BETTS
University of Rochester School of Medicine, Rochester, NY, ADMA Biologics, Hackensack, NJ

Abstract

Background: Respiratory Syncytial Virus (RSV) causes fatal respiratory tract infection in immunocompromised patients. Infusion of immunoglobulin (IG) with high levels of neutralizing RSV antibody (RSV-IVIG) may offer therapeutic benefit in these patients.
Methods: Compassionate use of RSV IVIG (RI-001-RSV-IVIG, ADMA Biologics) was allowed in 3 immunocompromised patients from the US [orthotopic liver transplant, acute myelogenous leukemia and chronic lymphocytic leukemia (CLL)] and 4 from Australia with documented RSV lower respiratory tract infection. RSV-IVIG was administered at 1500mg/kg on day 1 and 750mg/kg on day 3. Patients were also treated with inhaled ribavirin. Patients 1 & 2 received standard IVIG 2 days before RSV-IVIG. RSV antibody titers were measured by microneutralization assay pre and post each infusion. RSV RNA was quantified in respiratory secretions of the patient with CLL.
Results: RSV-IVIG was without serious side affects. All recovered and all had ~ 4-fold or greater rises in RSV-neutralizing antibody. The patient with CLL had no measurable antibody pre-infusion and a GMT of 7700 after the second infusion. Associated with the rise in serum antibody was a 10-fold drop in viral RNA in sputum from pre-infusion to day 4 with complete clearing of respiratory symptoms and normalization of oxygenation. All 7 patients developed titers of $\geq$2500 post infusion.
Conclusion: Administration of high titer RSV immunoglobulin was associated with an increase in serum neutralizing antibodies in seven immunocompromised adults with RSV lower respiratory tract infection. Future studies are needed to determine efficacy.

Background

Immunocompromised patients are at high risk for serious sequelae due to RSV infection. Patients with hematologic malignancies,solid organ and hematopoetic stem cell transplant (HSCT) recipients are particularly susceptible to fulminant infection. The reported mortality rate in pre-engraftment HSCT patients who develop RSV-LRTI can be as high as 70-100% without treatment. Inhaled Ribavirin and standard IVIG have been used in the treatment of RSV disease with some success; however, morbidity and mortality remain high in this patient population. RespiGam® (high titer RSV IVIG) was first introduced in the United States in 1996, but was voluntarily withdrawn from the market by the manufacturer in December 2003 after the introduction of Palivizumab. Because Palivizumab is dosed by weight, its use in adults is prohibitively expensive.

RSV-IVIG (RI-001)

The product, manufactured by Adma Biologics, is an aqueous preparation of immunoglobulins obtained from pooled plasma from donors with naturally occurring high titers of RSV as measured by microneutralization assay. The pooled plasma containing high-titers of anti-RSV antibodies was concentrated and treated with solvent/detergent to eliminate enveloped viruses. RI-001 is similar to RespiGam®.

Compassionate Use RI-00I
The safety and efficacy of RI-00I is presently being evaluated in a clinical trial of immunocompromised patients with documented RSV upper respiratory tract infection who are at risk for progression to lower tract disease (LRI). Patients with LRI at presentation were excluded from the study but eligible for compassionate use of the product on a case by case basis with application to the FDA for emergency IND. Seven patients received compassionate use treatment. RSV-IVIG was administered at 1500mg/kg on day 1 and 750mg/kg on day 3. Serum was collected pre and post infusion on days 1 and 3, as well as days 8, 10, 18 and 33. Five of seven patients also received inhaled ribavirin.

Microneutralization Assay
Serial dilutions of serum was mixed with a standard amount of A2 (A strain) RSV for 30 minutes in microtiter plates. HEp-2 cells were then added to the wells and the plates incubated at 37ºC in CO2 for 3 days. The plates were fixed and RSV antigen quantified by EIA using an RSV monoclonal antibody. The neutralization titer was defined as the titer of serum that reduces color development by 50%.

RSV Neutralizing Antibody Titers in Seven Compassionate Use Patients

	US Patients			Australian Patients			
Patient initials	**1**	**2**	**3**	**4**	**5**	**6**	**7**
Day 1 Pre-dose	362	1448	51	322	483	241	207
Day 1 Post-dose	2896	3861	7723	5148	3861	3861	4965
Day 3 Pre-dose	2896	3861	3861	1287	1931	3861	1241
Day 3 Post-dose	2896	5792	3861	2574	7723	11584	4965
Day 8		5792	3861	5148	5148		2482
Day 10			1931			7723	
Day 18		11584	965	1287	1931	3861	1655
Day 33				965	2574	2896	

Fold Rise in Antibody Titers following RSV-IVIG

	US Patients			Australian Patients			
Time Point	**1**	**2**	**3**	**4**	**5**	**6**	**7**
Day 1 Post-Treatment	8	3	151	16	8	16	24
Day 3 Pre-Treatment	8	3	76	4	4	16	6
Day 3 Post-Treatment	8	4	76	8	16	48	24
Day 8		4	76	16	11		12
Day 10			38			32	
Day 18		8	19	4	4	16	8
Day 33				3	5	12	

CASE REPORT - Subject 3

The patient is a 44 yo man with CLL undergoing intensive chemotherapy. On 4/5/09 the patient developed a URI. He was initially febrile with nasal and ear symptoms which slowly resolved but he developed progressive dyspnea and a productive cough after 10 days of illness. Chest CT showed diffuse bronchiolitis. He was seen in the infectious disease clinic on 4-15-09 and RT-PCR of his sputum sample was positive for a high titer of RSV group B. He was pale and dyspneic with diffuse wheezes and rales on lung exam. Oxygen saturation dropped from 97% to 93% on RA during minimal exercise. Because he remained ill and required additional chemotherapy in the near future he was admitted to the hospital on 4-17-09 for treatment. On 4-17-09 he received RSV IVIG at 1500mg/kg (day 1) and 750mg/kg on day 3. By day 4 his wheezing had resolved and was feeling much improved. Oxygenation normalized and he was discharged on day 5. He was seen in clinic on 4/24, 4/27 and 5/1 and was doing well. The quantity of RSV RNA diminished over time and became negative on 5/1. The patient began chemotherapy for his CLL the week of 5/7 and has had no recurrence of RSV.

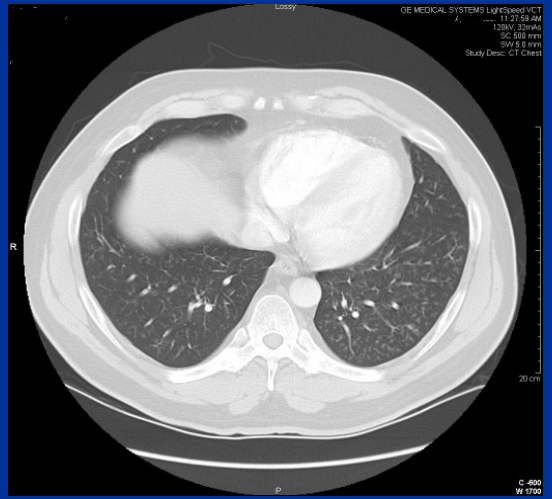

RSV Bronchiolitis on CT Scan

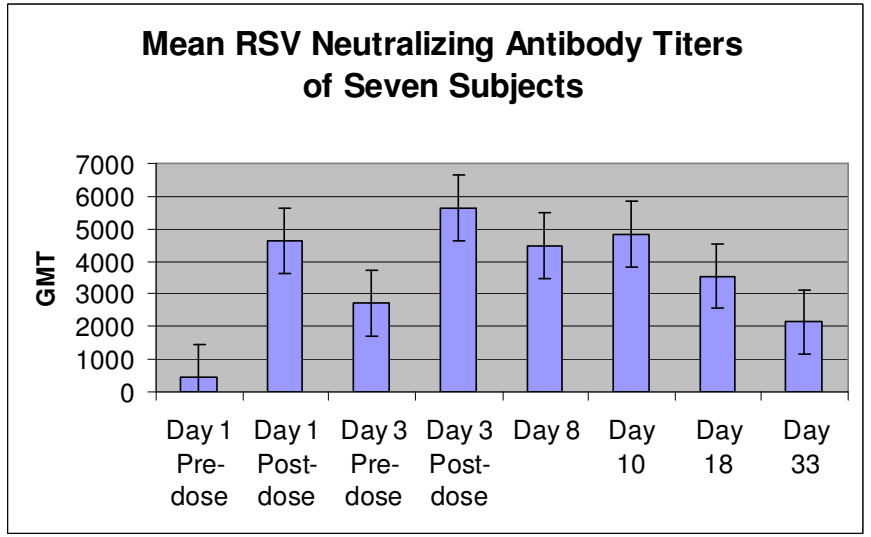
Mean RSV Neutralizing Antibody Titers
of Seven Subjects
GMT
7000
6000
5000
4000
3000
2000
1000
0
Day 1 Pre-dose
Day 1 Post-dose
Day 3 Pre-dose
Day 3 Post-dose
Day 8
Day 10
Day 18
Day 33

Patient Diagnoses and Outcomes

Patient	Age	Underlying Disease	RSV Diagnosis	Outcome	Serious Adverse Events
1	59 yo	Liver Transplant	Pneumonia	Survived	None
2	67 yo	Acute Myelogenous Leukemia	Pneumonia	Survived	None
3	44 yo	Chronic Lymphocytic Leukemia	Bronchiolitis	Survived	None
4	2 yo	HSCT	Pneumonia	Survived	None
4	8 yo	HSCT	Pneumonia	Survived	None
6	46 yo	HSCT with GVHD	Pneumonitis	Survived	None
7	9 yo	ALL sp cord transplant	Pneumonia	Survived	None

Conclusion

Administration of RSV-IVIG (RI-001) was associated with significant increases in serum neutralizing antibody titers in 7 immunocompromised patients and was associated with good clinical outcomes. Further study of high titer RSV-IVIG is warranted.