Locally secreted Fc-OX40L is superior to systemic, antibody mediated, OX40 co-stimulation for combination immunotherapy

George Fromm, Jason Rose and Taylor H. Schreiber
Heat Biologics, Inc., Durham NC

Abstract

The clinical success of checkpoint inhibitory therapy in a small percentage of patients has highlighted the need to identify combination approaches that may increase the frequency of responders. Two immunotherapy modalities that are proposed to synergize both with each other, and with checkpoint inhibitors are therapeutic vaccines and T cell co-stimulators.

To identify which T cell co-stimulators enhance the efficacy of an allogeneic, gp96-Ig secreting, cell-based vaccine (*Im*PACT), we investigated the activity of agonistic antibodies targeting OX40, 4-1BB and ICOS administered together with *Im*PACT. These data demonstrated that antigen-specific CD8+ T cell expansion is significantly enhanced by OX40, but not 4-1BB or ICOS stimulation.

Since T cell co-stimulation occurs at the site of immunization, we asked if co-expression of Fc-OX40L by the gp96-Ig secreting allogeneic vaccine cells (**new vaccine: *Com*PACT**) would provide comparable co-stimulation to systemically administered OX40 agonist antibodies. Interestingly, these data demonstrated that locally secreted Fc-OX40L by *Com*PACT provided superior priming of antigen-specific CD8+ T cells (peak of 13.3% of total CD8+) compared to combinations with OX40 antibodies (8.4%) or vaccine alone (5.6%).

Improved response was related to more potent activation of $CD127^{+}KLRG\text{-}1^{-}$ memory precursor cells by *Com*PACT. Systemic administration of OX40 antibodies also led to proliferation of non-specific CD4+ T cells, Tregs and systemic increases in IL-4, IL-5, IL-6, TNFα and IFNγ. Importantly, *Com*PACT led to high frequencies of $IFN\gamma^{+}$, $TNF\alpha^{+}$, granzyme-b^{+} and IL-2^{+} antigen-specific CD8+ T cells at both priming and boosting, which enhanced rejection of established CT26 tumors.

These data demonstrate that vaccination and co-stimulation can be achieved with a single cell-based product, which may simplify clinical development by enhancing the activation of tumor-antigen specific CD8+ T cells.

Gp96-Ig Vaccine and T Cell Co-stimulator Synergy

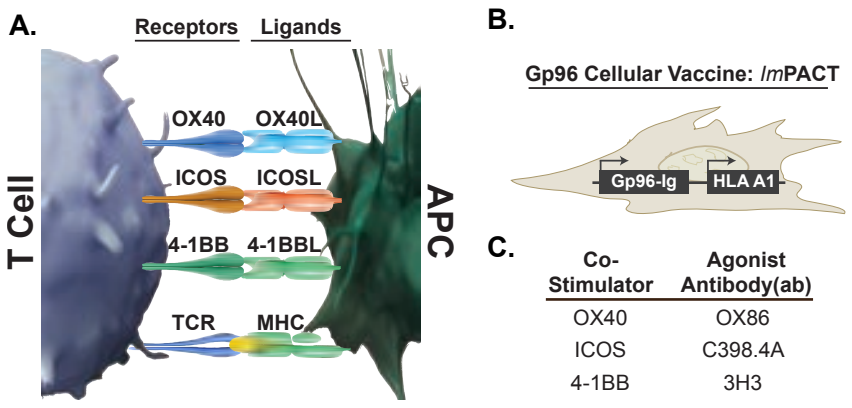
A.
Receptors
Ligands
T Cell
APC
OX40
OX40L
ICOS
ICOSL
4-1BB
4-1BBL
TCR
MHC
B.
Gp96 Cellular Vaccine: ImPACT
Gp96-Ig
HLA A1
C.

Co-Stimulator	Agonist Antibody(ab)
OX40	OX86
ICOS	C398.4A
4-1BB	3H3

Figure 1. Testing synergy between *Im*PACT and T cell co-stimulators. (A) Diagram of co-stimulator receptors and ligands on T cells and antigen presenting cells (APC). (B) Schematic of Gp96-Ig *Im*PACT vaccine. (C) Co-stimulator antibodies analyzed.

*Im*PACT Synergy with OX40, but not 4-1BB or ICOS Agonist mAbs

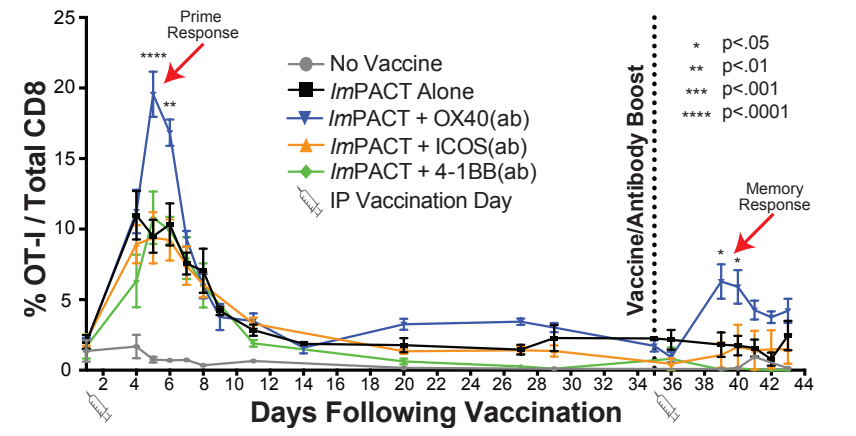
Prime Response
Memory Response
% OT-I / Total CD8
Days Following Vaccination
Vaccine/Antibody Boost
No Vaccine
ImPACT Alone
ImPACT + OX40(ab)
ImPACT + ICOS(ab)
ImPACT + 4-1BB(ab)
IP Vaccination Day
* p<.05
** p<.01
*** p<.001
**** p<.0001

Figure 2. OX40 antibody synergizes with gp96-Ig vaccine to produce T cell expansion. Mice transferred with OT-I (EGFP) cells via tail vein injection on day -1, were then vaccinated with *Im*PACT +/- T cell co-stimulator agonistic antibodies for OX40, ICOS and 4-1BB, and then analyzed by flow cytometry. Mice were boosted with the same combinations on day 35.

*Com*PACT : New Vaccine Combining Gp96-Ig with OX40L-Fc

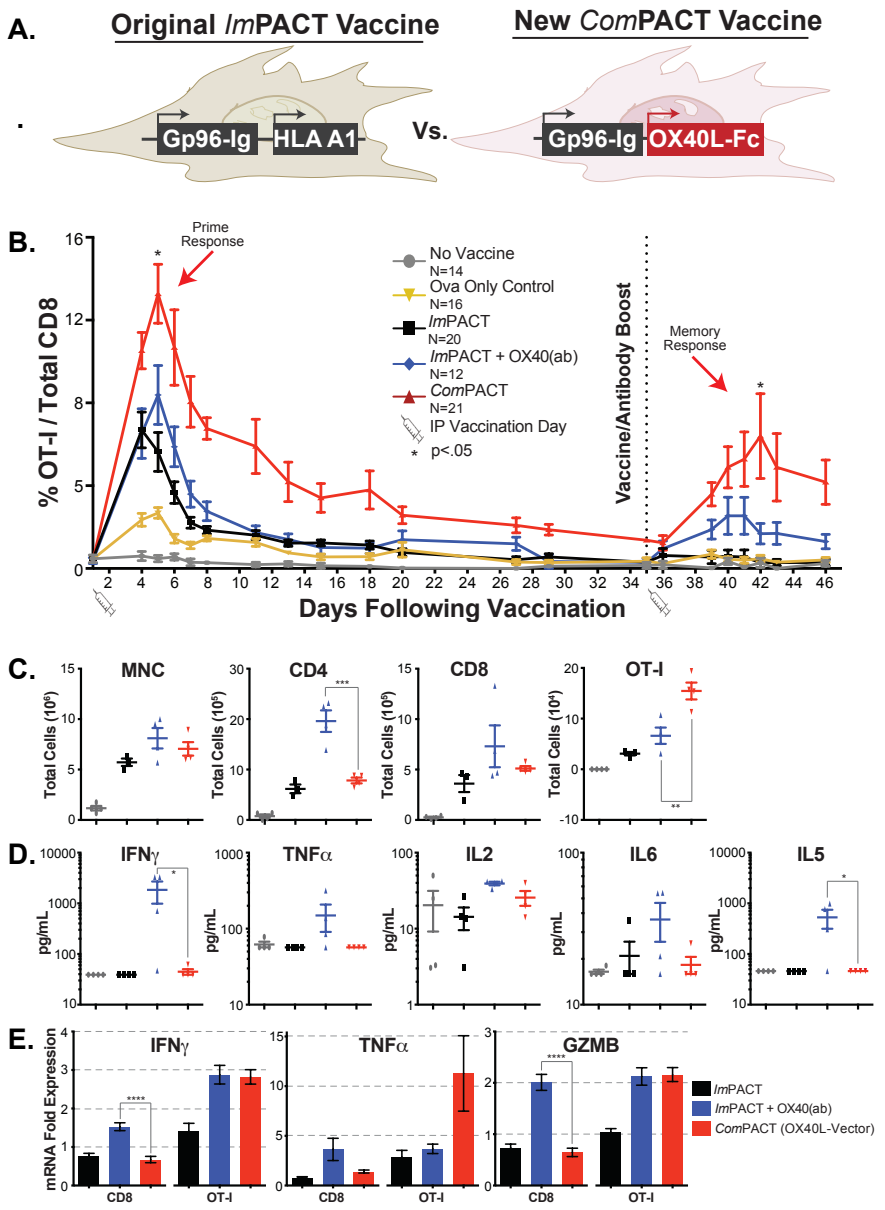
A.
Original ImPACT Vaccine
New ComPACT Vaccine
Gp96-Ig
HLA A1
Vs.
OX40L-Fc
B.
Prime Response
Memory Response
% OT-I / Total CD8
Days Following Vaccination
Vaccine/Antibody Boost
No Vaccine N=14
Ova Only Control N=16
ImPACT N=20
ImPACT + OX40(ab) N=12
ComPACT N=21
IP Vaccination Day
* p<.05
C.
MNC
CD4
CD8
OT-I
Total Cells
D.
IFNγ
TNFα
IL2
IL6
IL5
pg/mL
E.
mRNA Fold Expression
GZMB
ImPACT
ImPACT + OX40(ab)
ComPACT (OX40L-Vector)

Figure 3. ***Com*PACT generates greater T cell expansion than OX40 antibody, and is specific to antigen related CD8 cells.** (A) Schematic of new vaccine *Com*PACT. (B) Antigen specific (OT-I/ EGFP) CD8 T cell expansion analyzed by flow cytometry following vaccination and boost by *Im*PACT +/- OX40(ab) or *Com*PACT. Mice were analyzed at day 8 by (C) peritoneal flow cytometry, (D) blood serum cytokines and (E) T cell activation qRT-PCR on sorted CD8 only or OT-I cells. OX40(ab) results in non-specific global activation of immune response, compared to antigen specific CD8 response of *Com*PACT.

*Com*PACT Generates More MPEC Than OX40 Agonist mAbs

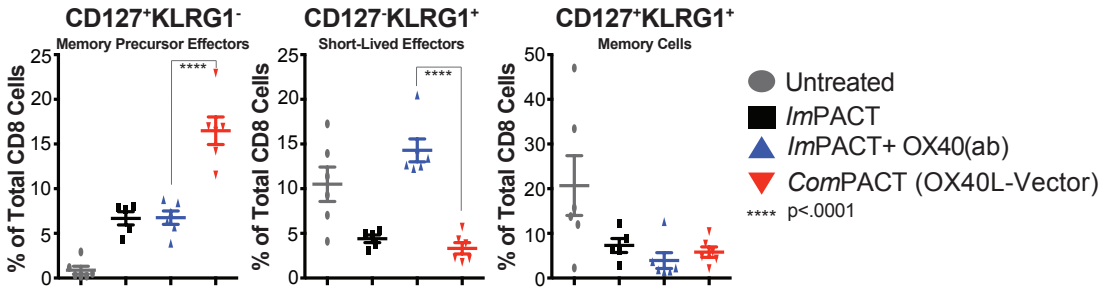
CD127+KLRG1-
Memory Precursor Effectors
CD127-KLRG1+
Short-Lived Effectors
CD127+KLRG1+
Memory Cells
% of Total CD8 Cells
Untreated
ImPACT
ImPACT+ OX40(ab)
ComPACT (OX40L-Vector)
**** p<.0001

Figure 4. *Com*PACT generates potent Memory Precursor Effector Cell (MPEC) activation allowing for robust immune response after boost. CD8 cells were analyzed by flow cytometry on day 8 of the time-course described in figure 3.

Survival Benefit with *Com*PACT and OX40 Agonist mAbs

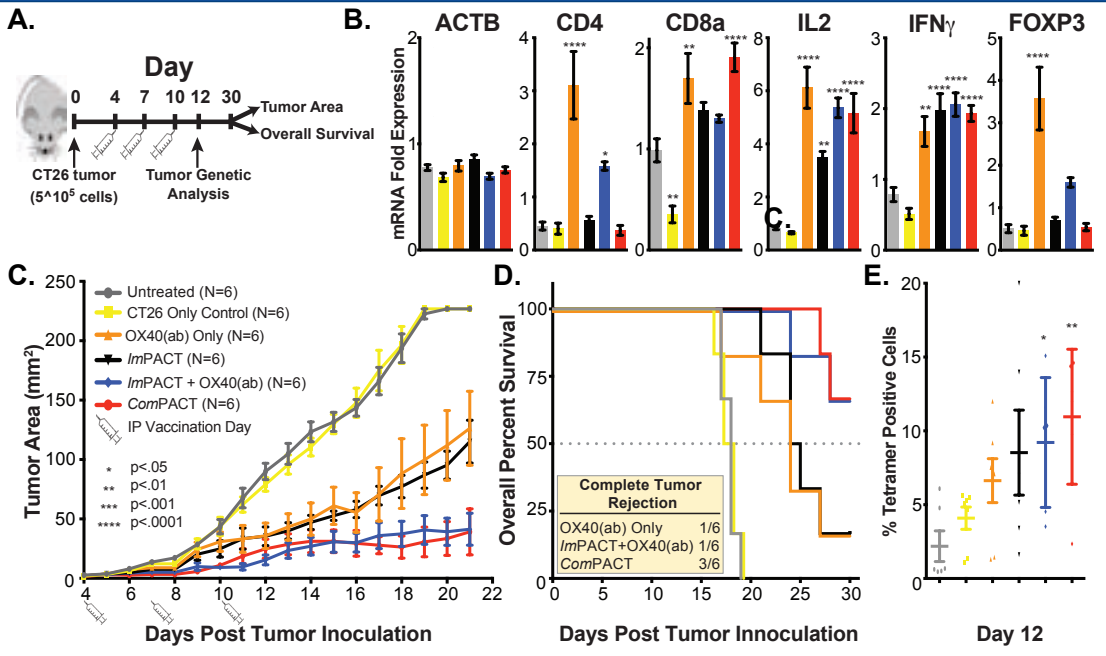
A.
Day
0 4 7 10 12 30
Tumor Area
Overall Survival
CT26 tumor (5^10^5 cells)
Tumor Genetic Analysis
B.
ACTB
CD4
CD8a
IL2
IFNγ
FOXP3
mRNA Fold Expression
C.
Untreated (N=6)
CT26 Only Control (N=6)
OX40(ab) Only (N=6)
ImPACT (N=6)
ImPACT + OX40(ab) (N=6)
ComPACT (N=6)
IP Vaccination Day
Tumor Area (mm²)
Days Post Tumor Inoculation
D.
Overall Percent Survival
Complete Tumor Rejection
OX40(ab) Only 1/6
ImPACT+OX40(ab) 1/6
ComPACT 3/6
Days Post Tumor Innoculation
E.
% Tetramer Positive Cells
Day 12

Figure 5. *Com*PACT treatment results in antigen specific CD8+ tumor infiltration, block in tumor growth, increase in survival and significant tumor rejection. (A) Experimental setup: tumor inoculation of $5x10^5$ CT26 cells on day 0 and vaccination on days 4, 7 and 10 ($1x10^6$ cells and 100 μg of antibody for appropriate treatments). (B) Day 12 genetic analysis of tumor isolated RNA. (C) Tumor growth over a 21 day time course. (D) Overall survival of mice in the various treatment groups. (E) CT26-specific AH1 antigen tetramer is significantly higher in *Com*PACT treated mice. Day 12 CD8+ splenocytes were negatively selected for CD11b, CD11c, Gr-1 and NK1.1, then positively selected for CD8a and CD3, and finally the percent of AH1-tetramer+ cells was determined.

Statistical Analysis. One-way ANOVA was used for all sample group analyses. Significance is denoted by *, signifying the following: *p<.05, **p<.01, ***p<.001, and ****p<.0001. Sample sizes are noted in experiments and represent a minimum of 3 distinct biological replicates with error as SEM.

Key Concepts

-We have developed a novel, next-generation cancer immunotherapy vaccine to Gp96-Ig, which we call ***Com*PACT**, incorporating T cell co-stimulator Fc-OX40L.

-*Com*PACT stimulates higher frequency proliferation of antigen-specific CD8+ T cells at both priming and boosting, and more MPEC, than OX40 agonist antibodies.

-*Com*PACT demonstrates greater antigen specificity, without off-target proliferation and systemic inflammatory cytokine stimulation seen with OX40 agonist mAbs.

-*Com*PACT delivers a vaccine and co-stimulatory fusion protein in a single compound, with superior specificity than traditional antibodies. This product may simplify the development of combination immunotherapeutics for oncology patients.

Contact: gfromm@heatbio.com

PhDPosters.com