P1174

Plecanatide, a Guanylate Cyclase-C Agonist, Improves Bowel Habits and Symptoms Associated with Chronic Constipation in a Phase IIa Clinical Study

Kunwar Shailubhai, Laura Barrow, Craig Talluto, Stephen Comiskey, John Foss, Rong Feng, Alan Joslyn, and Gary Jacob

Synergy Pharmaceuticals, Inc; 420 Lexington Ave, New York, NY 10170 & 1609 Old Easton Rd, Doylestown, PA 18902

Background

ABSTRACT

Uroguanylin and guanylin are physiological agonists of the guanylate cyclase-C (GC-C) receptor. Activation of the GC-C receptor stimulates intracellular synthesis of cyclic GMP, resulting in activation of the cystic fibrosis transmembrane conductance regulator (CFTR). Activation of CFTR results in augmented flow of fluid and bicarbonate into the intestinal lumen. An optimum volume of fluid secretion in the proximal intestine is critical for normal bowel movement and also for complete defecation. Thus, oral treatment with a GC-C agonist is expected to promote spontaneous bowel movements (SBMs) and to reduce abdominal discomfort. Plecanatide is a superior analog of uroguanylin. This study evaluated the safety and effectiveness of once daily plecanatide in patients with chronic constipation (CC).

Plecanatide was safe, well tolerated, and it improved bowel habits when compared with placebo across all doses. Moreover, the effects tended to be rapid and sustained. A large multicenter trial is planned to study plecanatide in CC patients to further evaluate efficacy and safety.

Mechanism of Action of GC-C agonists in the GI Tract

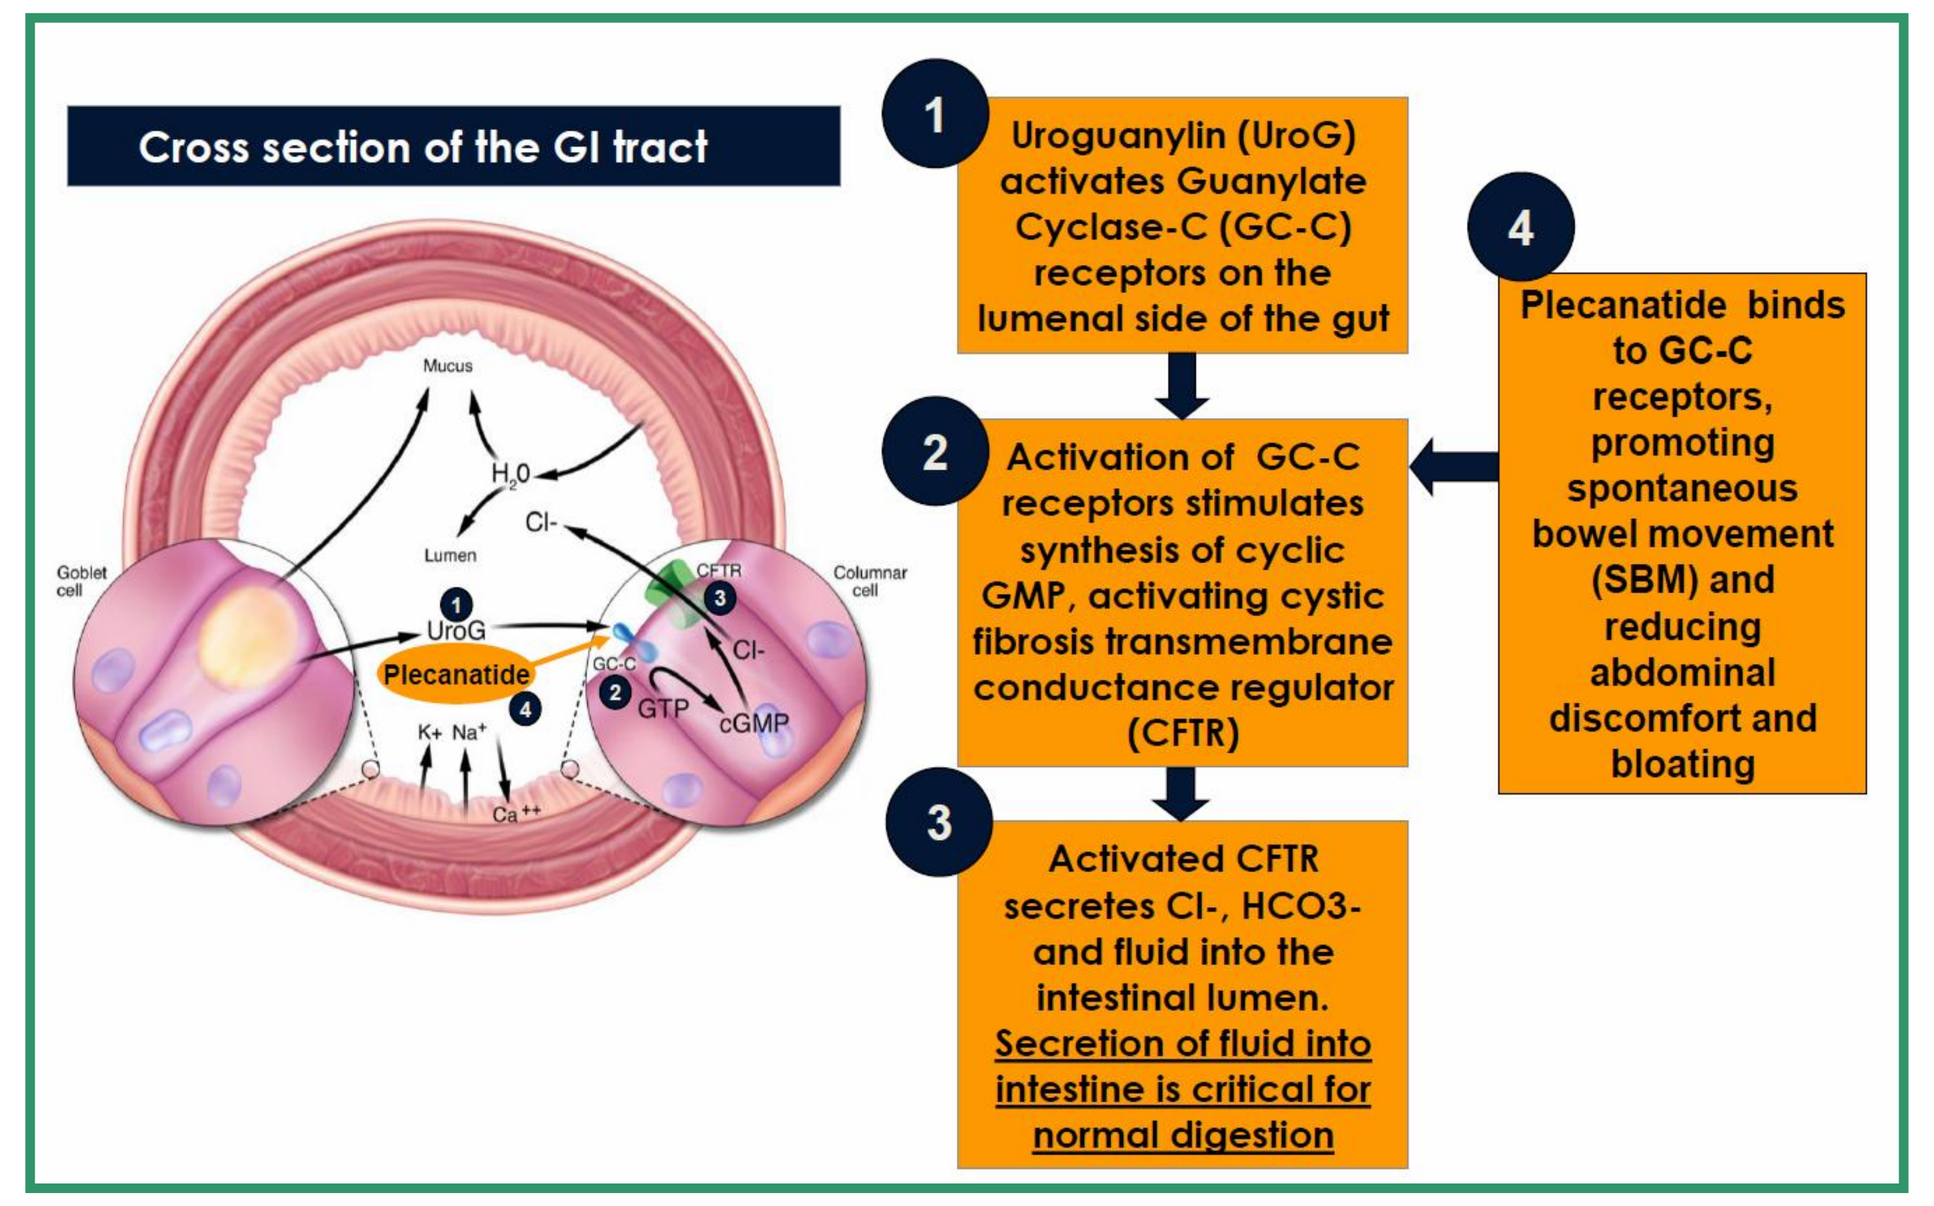

Cross section of the GI tract
Mucus
H_2O
Cl-
Lumen
Goblet cell
Columnar cell
UroG
Plecanatide
CFTR
GC-C
GTP
cGMP
K+ Na+
Ca++
1
Uroguanylin (UroG) activates Guanylate Cyclase-C (GC-C) receptors on the lumenal side of the gut
2
Activation of GC-C receptors stimulates synthesis of cyclic GMP, activating cystic fibrosis transmembrane conductance regulator (CFTR)
3
Activated CFTR secretes Cl-, HCO3- and fluid into the intestinal lumen. Secretion of fluid into intestine is critical for normal digestion
4
Plecanatide binds to GC-C receptors, promoting spontaneous bowel movement (SBM) and reducing abdominal discomfort and bloating

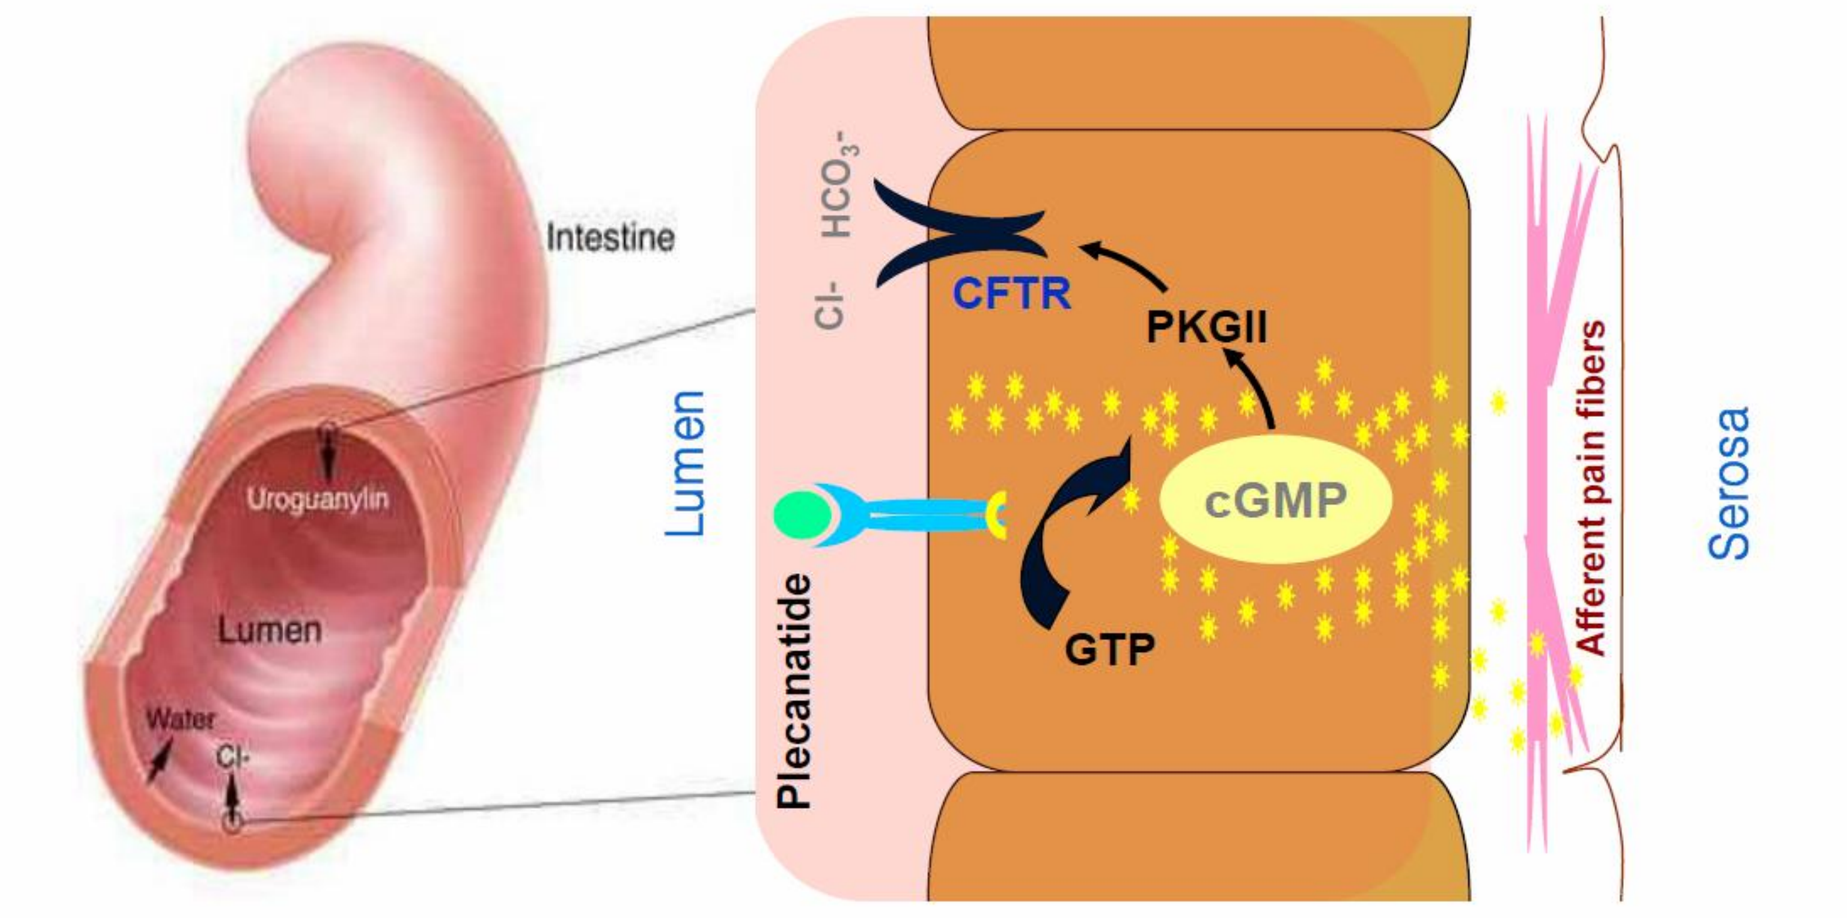

Intestine
Uroguanylin
Lumen
Water
Cl-
Lumen
Plecanatide
Cl-
HCO_3^-
CFTR
PKGII
cGMP
GTP
Afferent pain fibers
Serosa

CC: Abdominal discomfort due to fecal blockage and pressure

IBS-c: Abdominal pain due to low grade inflammation

Objectives

Primary Objective:

- To evaluate the safety of plecanatide for 14 days in patients with CC

Secondary Objectives:

- To assess changes in bowel habits over time (frequency, completeness of evacuation, stool consistency, straining and abdominal discomfort) during daily dosing with plecanatide or placebo.
- To assess the time to first bowel movement after first daily dose of plecanatide as compared with placebo
- To assess the pharmacokinetic (PK) profile after the first daily dose of plecanatide and prior to and after the last dose of plecanatide

Methods

Study Design

- This was a Phase IIa, randomized, double-blind, placebo-controlled, 14-day repeat, oral, dose-ranging study
- There were four dose cohorts in this trial:
 - Plecanatide (0.3 mg, 1.0 mg, 3.0 mg or 9.0 mg) and matching placebo
 - 3:1 randomization (plecanatide : matching placebo)
 - 20 subjects per dose cohort (15 plecanatide:5 matching placebo)
- Subjects took one daily dose in the morning for 14 days
- Blood samples for PK were drawn prior to dosing on days 1 and 14 and at 0.5, 1.0 and 2 hour post-dosing.
- After each cohort was completed, a safety review was conducted prior to initiating the next cohort

Primary Subject Selection Criteria:

- Meet modified ROME III criteria for CC
- Colonoscopy within past 5 years with no significant finding
- Good health as determined by physical examination, medical history, vital signs, ECG, clinical chemistry, hematology, urinalysis, drug screen and serology assessments
- During 14-day pretreatment period, subjects must report < 6 SBM and < 3 CSBM in each pre-treatment week

Table 1: Summary of Subject Demographics

	Placebo	0.3 mg	1.0 mg	3.0 mg	9.0 mg
Age*	47.7 (14.6)	51.1 (12.0)	50.5 (10.6)	48.5 (16.2)	47.3 (12.7)
Gender					
Female	18 (90.0%)	12 (85.7%)	14 (100%)	13 (86.7%)	12 (80%)
Male	2 (10.0%)	2 (14.3%)	0 (0%)	2 (13.3%)	3 (20%)
Race					
Caucasian	17 (85%)	13 (92.9%)	12 (85.7%)	14 (93.3%)	12 (80%)
African Am	1 (5%)	0	1 (7.1%)	0	2 (13.3%)
Asian Am	1 (5%)	1 (7.1%)	1 (7.1%)	0	1 (6.7%)
Am Indian	1 (5%)	0	0	0	0
Other	0	0	0	1 (6.7%)	0

* Mean (SD) Am = American

Methods

Statistical Analysis:

- Primary endpoints for the safety evaluation were summarized for the patients that received at least one dose of plecanatide or placebo (safety population)
- The secondary assessments for pharmacodynamic activity (efficacy) were summarized by descriptive statistics for the patients that received at least one post-baseline pharmacodynamic assessment [modified intent-to-treat population (equivalent to the safety population)]
 - Time to first bowel movement after first daily dose
 - Stool frequency (spontaneous bowel movements, SBMs)
 - Completeness of evacuation (complete spontaneous bowel movements, CSBMs)
 - Stool consistency (Bristol stool form scale, BSFS)
 - Straining (7-category scale)
 - Global assessments of abdominal discomfort, constipation severity, and overall relief (6-category scales)

Results – PK and Safety

Pharmacokinetics

- No detectable systemic absorption of plecanatide (assay sensitivity: 10 ng/mL)

Safety

- No deaths reported in this study
- No serious adverse events (SAE) reported in subjects receiving plecanatide
- % subjects who reported AEs considered related to treatment
 - Placebo 10% (2/20)
 - Plecanatide 17.2% (10/58)
- % subjects who reported GI-related AEs (related or unrelated to treatment)
 - Placebo 10% (2/20)
 - Plecanatide 8.6% (5/58)
- Majority of adverse events (AE) were mild / moderate and transient in nature
- No diarrhea reported for any subject receiving plecanatide
 - One subject on placebo discontinued from the study due to diarrhea

Table 2: Summary of All Adverse Events

Event	Placebo n=20	0.3 mg n=14	1.0 mg n=14	3.0 mg n=15	9.0 mg n=15
Total AE	4 (20.0%)	3 (21.4%)	6 (42.9%)	2 (13.3%)	6 (40.0%)
Treatment Emergent AE	4 (20.0%)	3 (21.4%)	6 (42.9%)	2 (13.3%)	6 (40.0%)
Severe AEs	0	1 (7.1%)	0	1 (6.7%)	3 (5.2%)
Serious AEs	1 (5.0%)	0	0	0	0
Related AEs	2 (10.0%)	2 (14.3%)	2 (14.3%)	1 (6.7%)	5 (33.3%)
AEs Leading to Discontinuation	1 (5.0%)	0	0	0	0

Results – Safety

Table 3: GI-Related Adverse Event Summary with Relationship to Treatment

AE	Placebo	0.3 mg	1.0 mg	3.0 mg	9.0 mg
Ab Cramping	1 (5.0%)	0	0	0	0
Ab Pain	1 (5.0%)	0	0	0	0
Bloating	0	0	0	0	1 (6.7%)
Diarrhea	1 (5.0%)	0	0	0	0
Flatulence	2 (10.0%)	0	0	0	0
Nausea	0	1 (7.1%)	0	0	
Upset Stomach	0	0	0	1 (6.7%)	0

Ab = Abdominal

Results - Efficacy

Median Time to First Bowel Movement after Start of Treatment

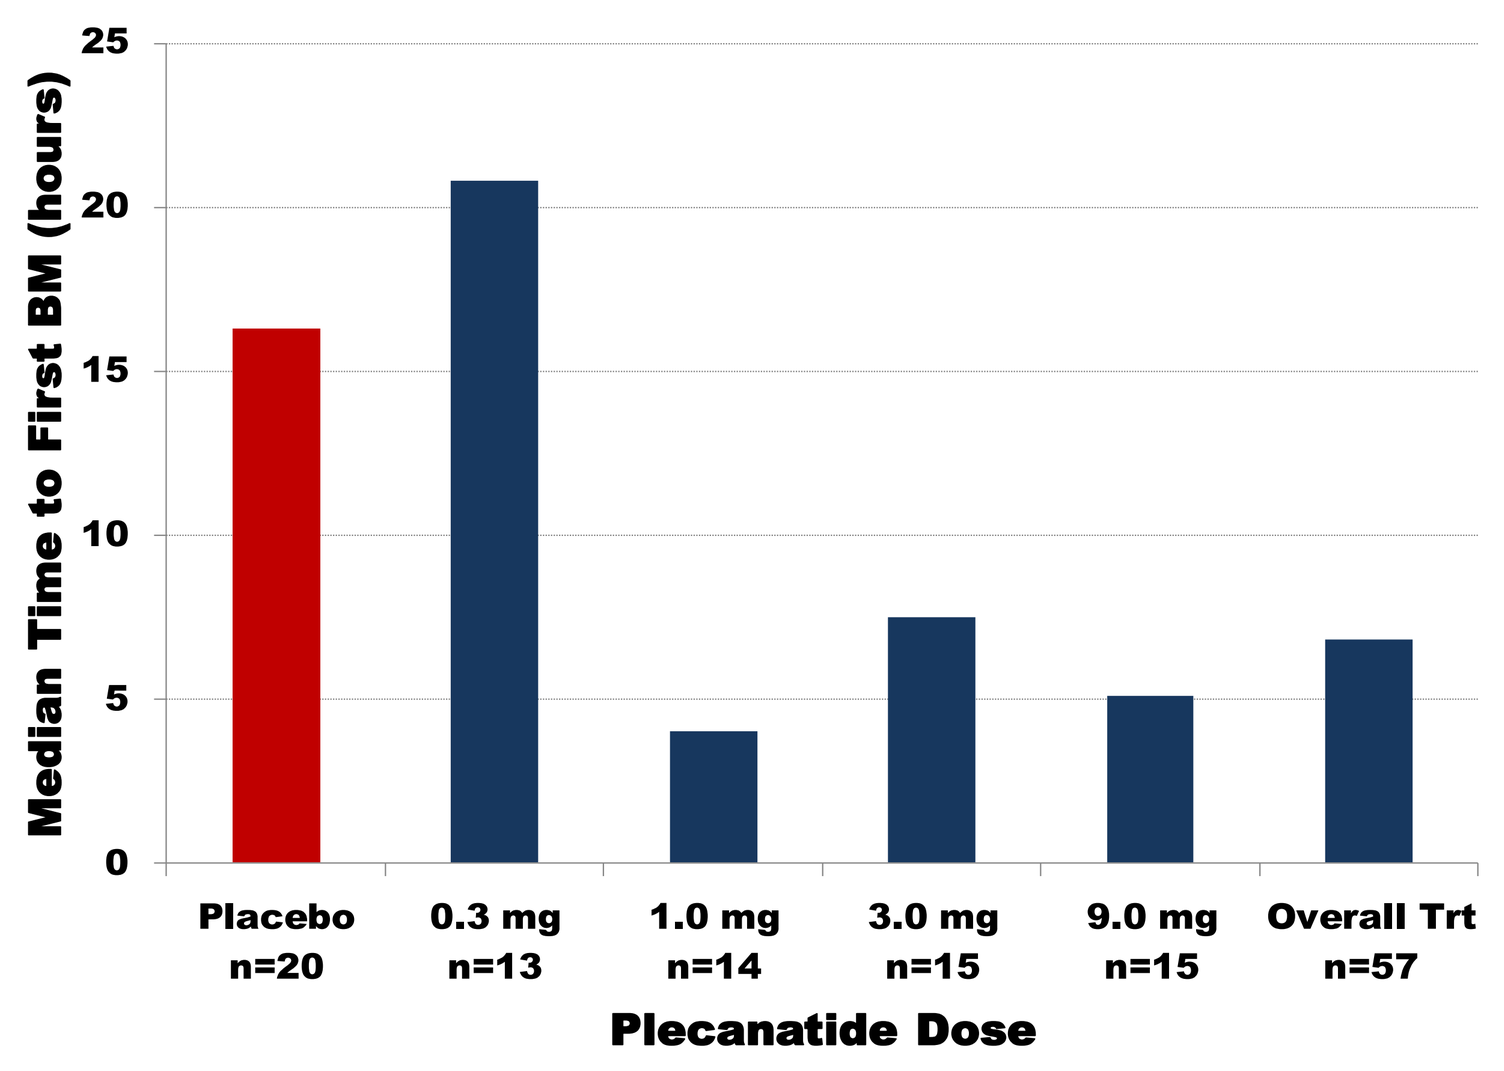

Median Time to First BM (hours)
25
20
15
10
5
0
Placebo n=20
0.3 mg n=13
1.0 mg n=14
3.0 mg n=15
9.0 mg n=15
Overall Trt n=57
Plecanatide Dose

Spontaneous Bowel Movements (SBMs) During Baseline and Treatment Periods

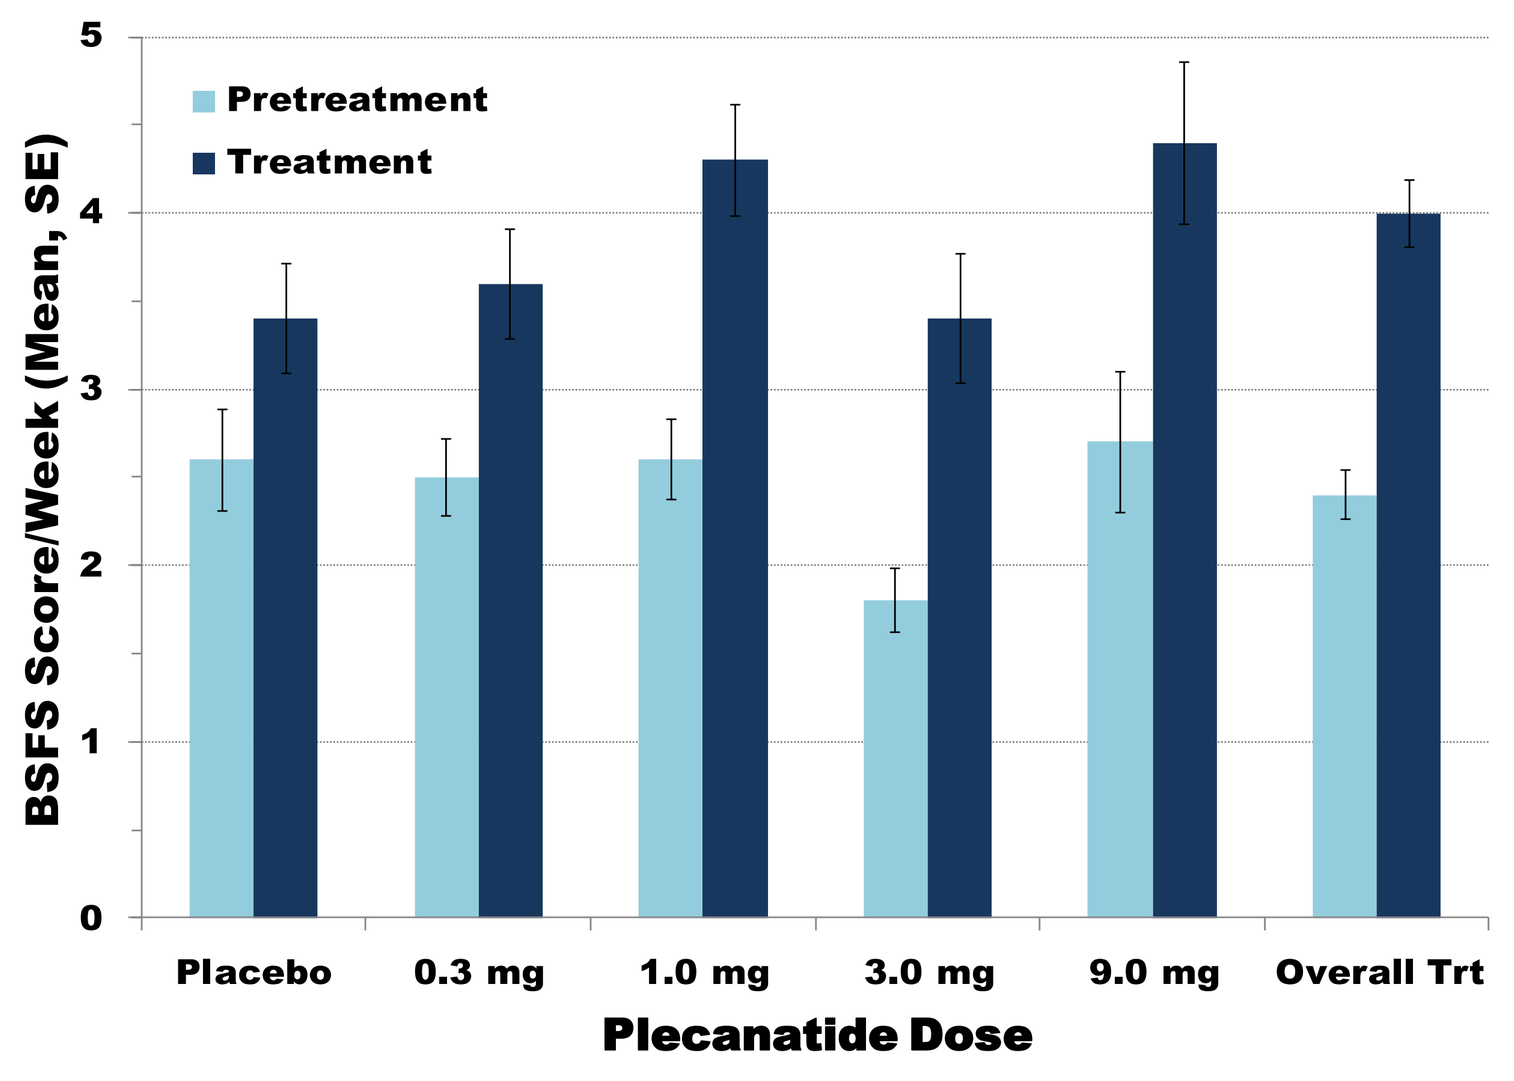

BSFS Score/Week (Mean, SE)
Pretreatment
Treatment
5
4
3
2
1
0
Placebo
0.3 mg
1.0 mg
3.0 mg
9.0 mg
Overall Trt
Plecanatide Dose

Results - Efficacy

Complete Spontaneous Bowel Movements During Baseline and Treatment Periods

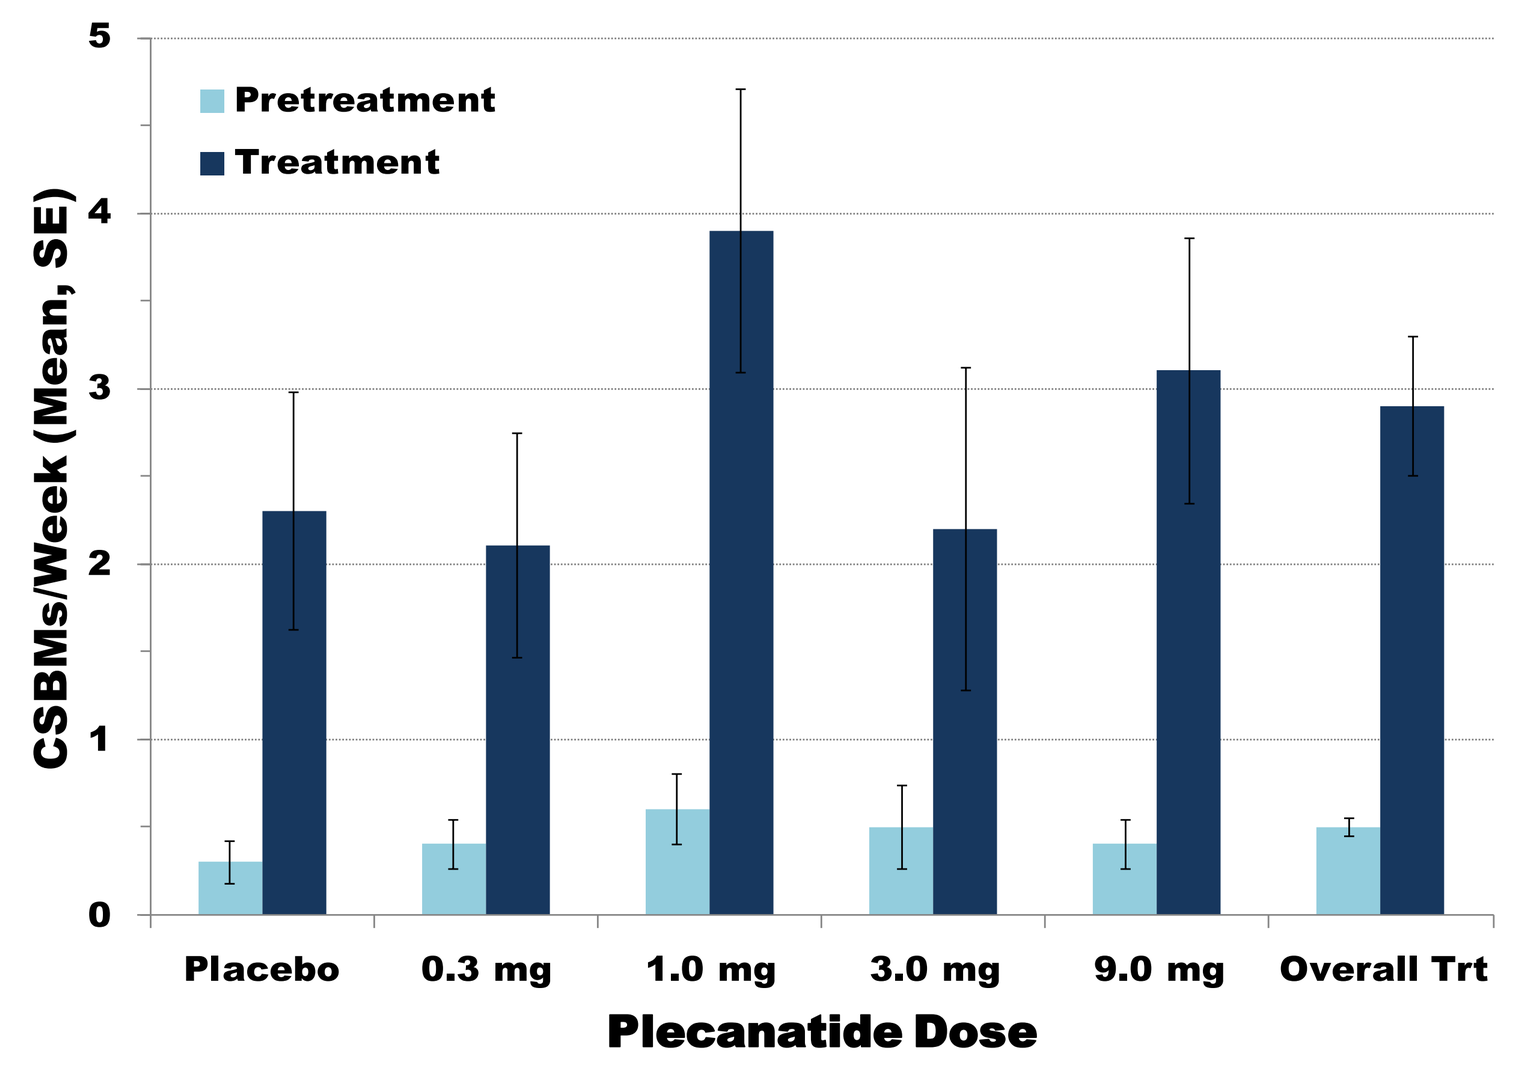

CSBMs/Week (Mean, SE)
Pretreatment
Treatment
5
4
3
2
1
0
Placebo
0.3 mg
1.0 mg
3.0 mg
9.0 mg
Overall Trt
Plecanatide Dose

Bristol Stool Form Score (BSFS) During Baseline and Treatment Periods

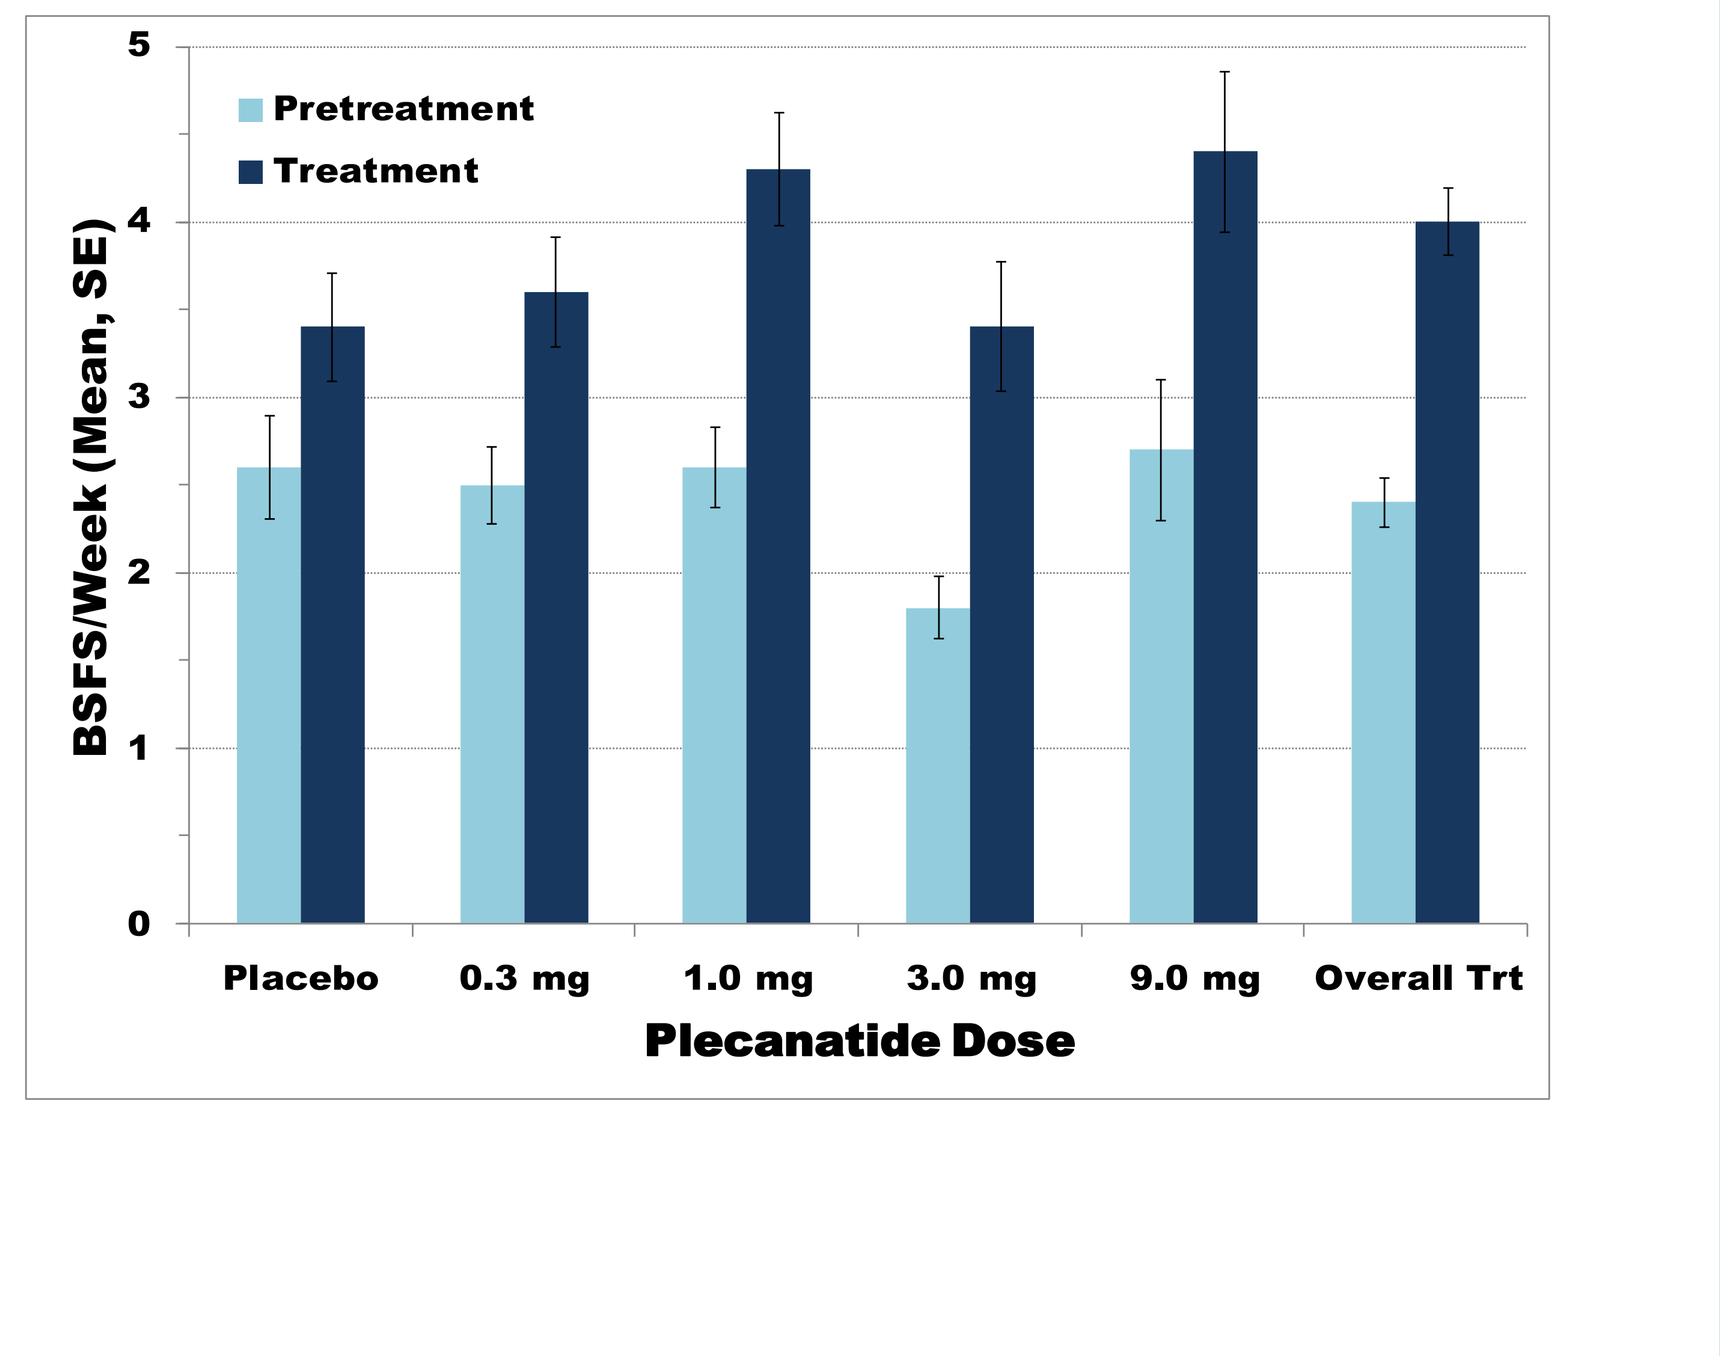

BSFS/Week (Mean, SE)
Pretreatment
Treatment
5
4
3
2
1
0
Placebo
0.3 mg
1.0 mg
3.0 mg
9.0 mg
Overall Trt
Plecanatide Dose

Straining Score During Baseline and Treatment Periods

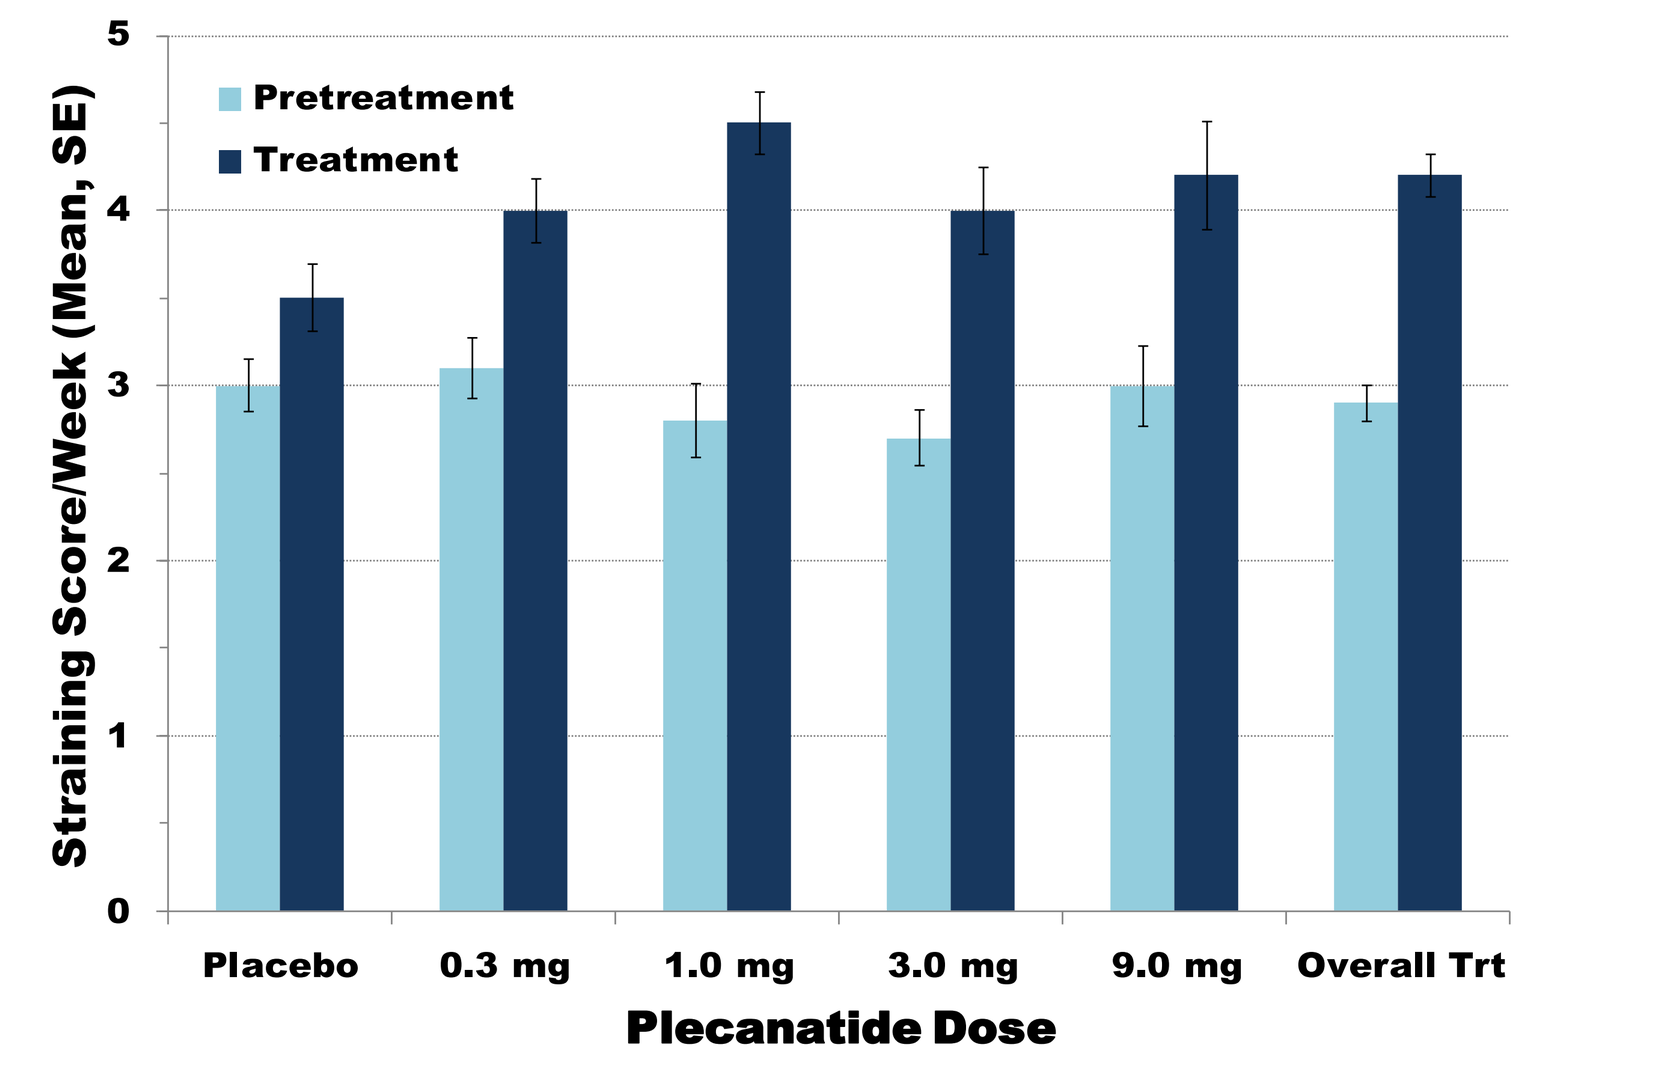

Straining Score/Week (Mean, SE)
Pretreatment
Treatment
5
4
3
2
1
0
Placebo
0.3 mg
1.0 mg
3.0 mg
9.0 mg
Overall Trt
Plecanatide Dose

Results - Efficacy

Global Assessment Scores after Treatment

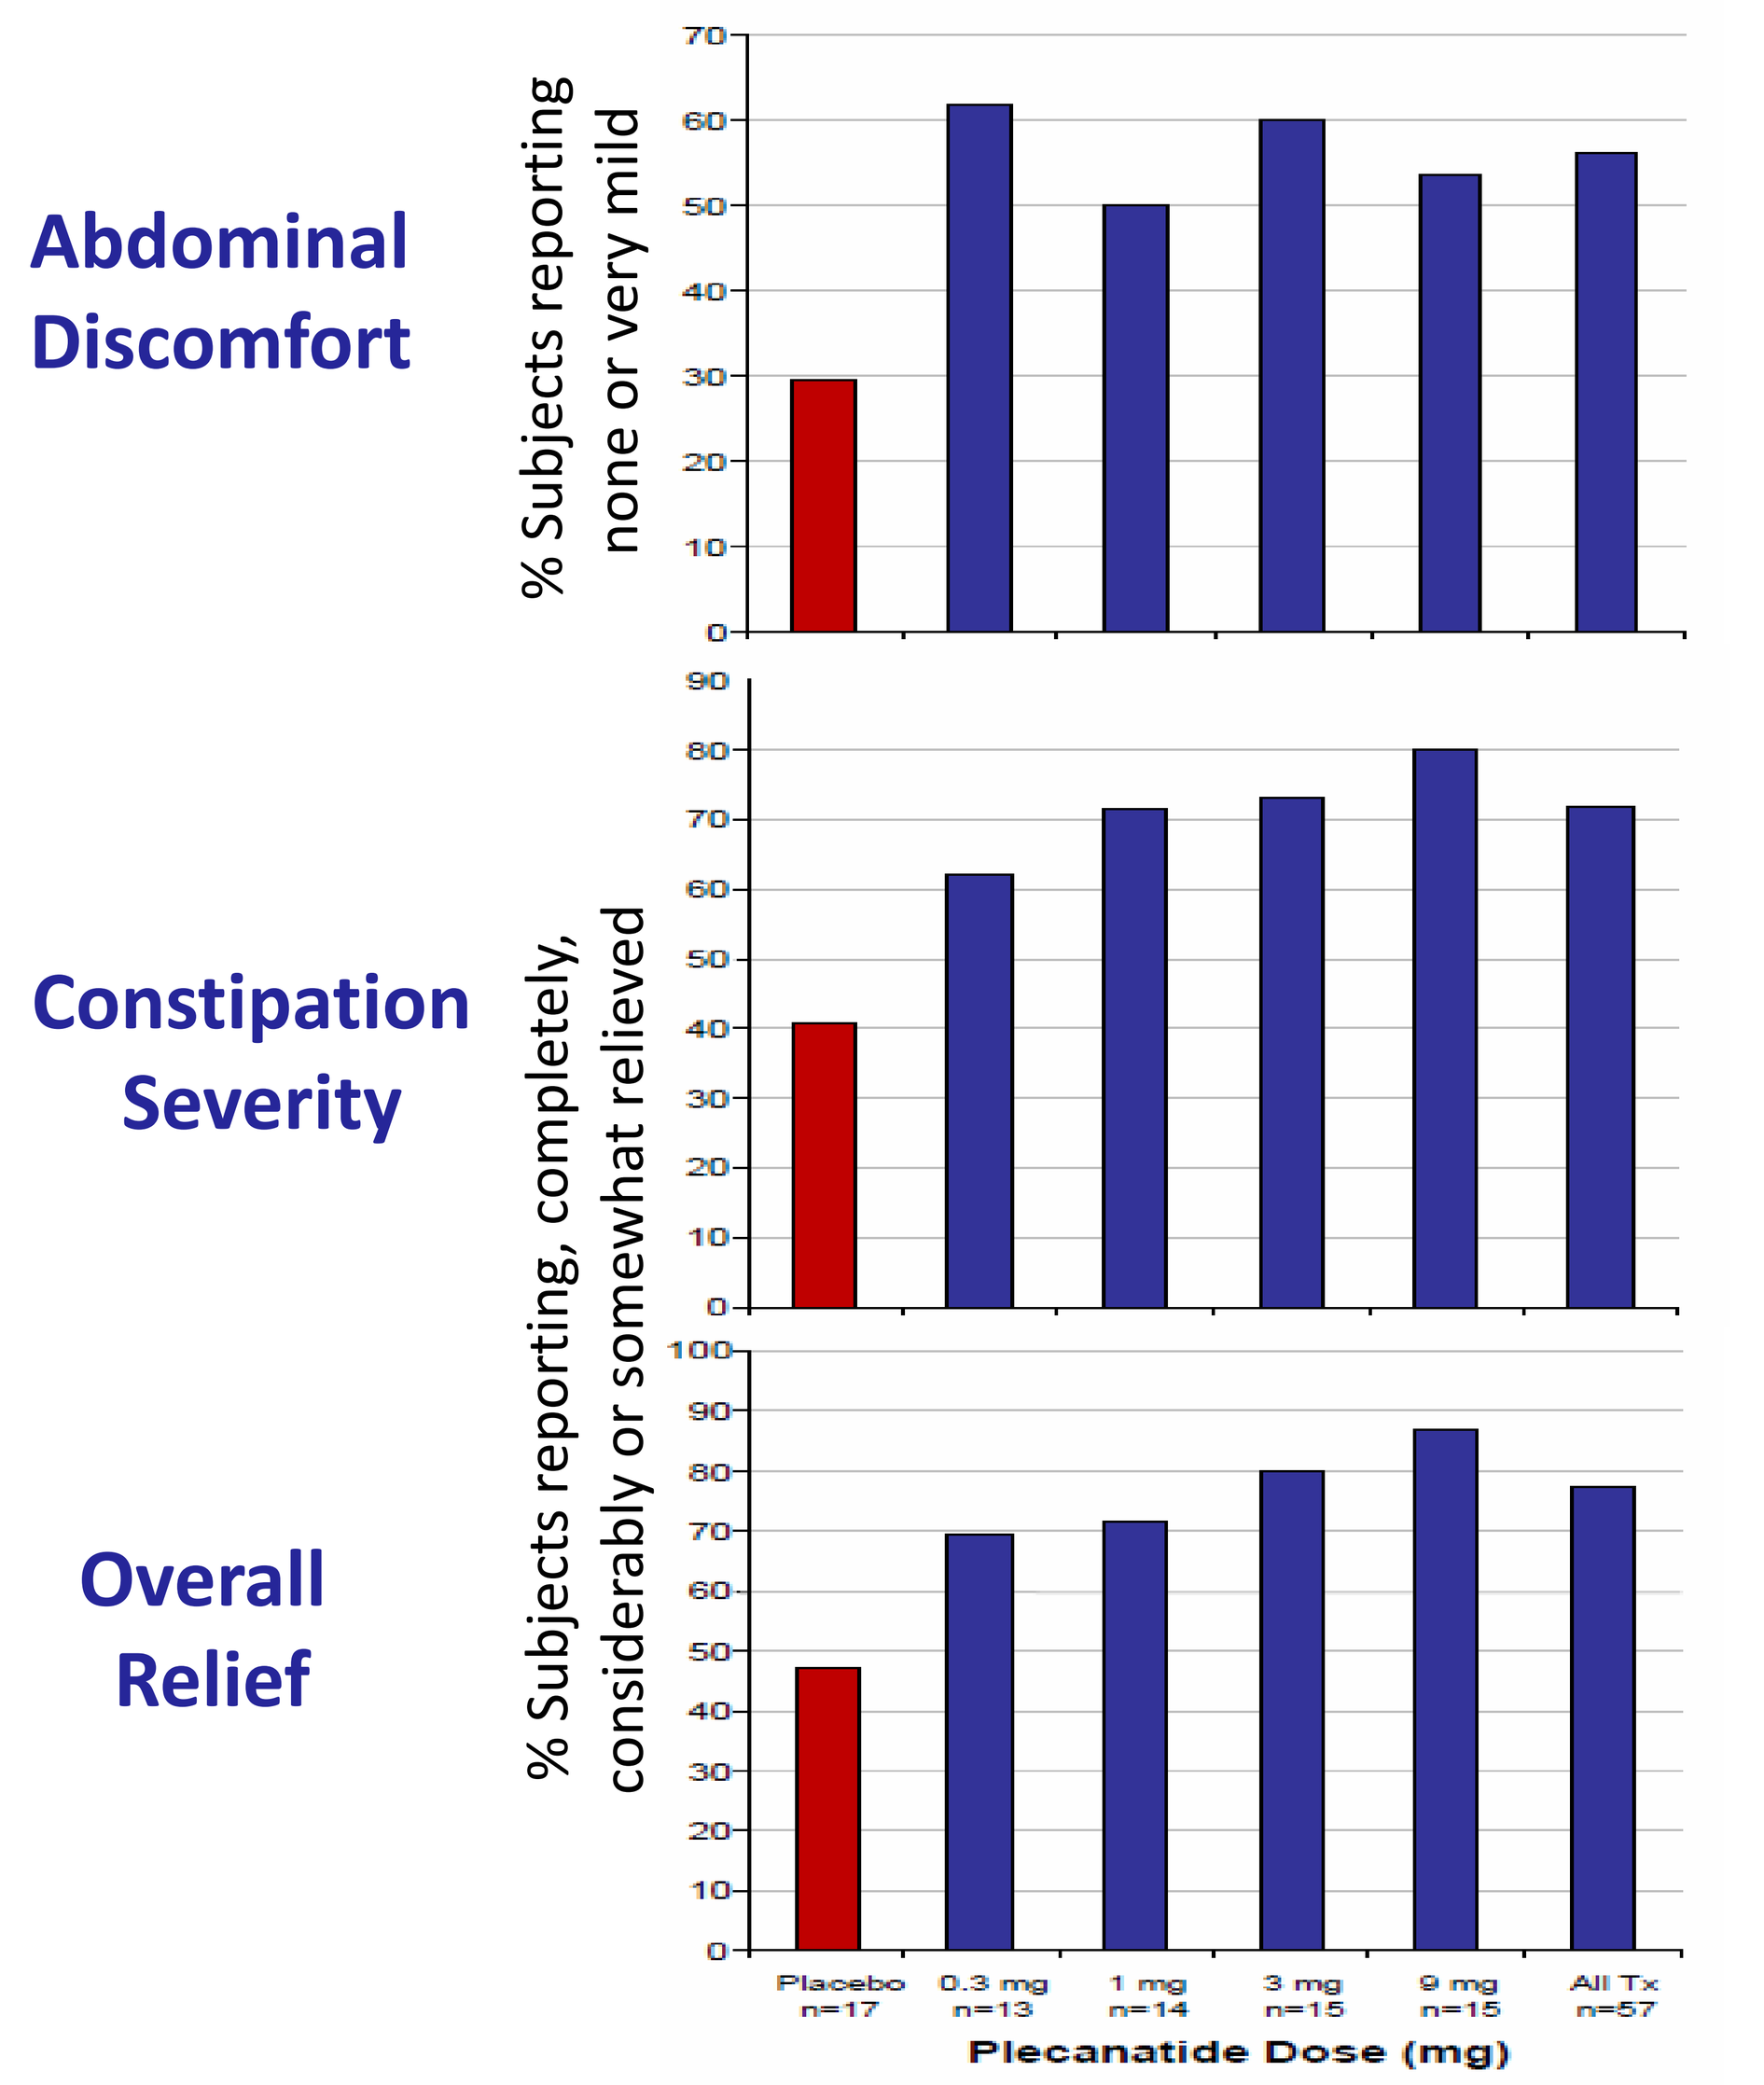

Abdominal Discomfort
Constipation Severity
Overall Relief
% Subjects reporting none or very mild
% Subjects reporting, completely, considerably or somewhat relieved
Placebo n=17
0.3 mg n=13
1 mg n=14
3 mg n=15
9 mg n=15
All Tx n=57
Plecanatide Dose (mg)

Conclusions

- There was no detectable systemic absorption of plecanatide at any dose up to 9 mg (assay sensitivity: 10 ng/mL)
- No SAEs reported in subjects receiving plecanatide
- The safety profile demonstrated minimal adverse events
- No reports of diarrhea in subjects who received plecanatide
- All plecanatide groups showed a significant decrease in time to first bowel movement
- Plecanatide demonstrated increases in stool frequency (SBM and CSBM)
- Plecanatide improved stool consistency, reduced straining and reduced abdominal discomfort

This information concerns an investigational agent that has not been approved by the US Food and Drug Administration.

Presented at the 2011 American College of Gastroenterology Annual Scientific Meeting in Washington DC, 28 Oct to 2 Nov.